ASCPT 2023, MARCH 22-24, ATLANTA, GA, USA

PI-059 Early Clinical PK/PD Modeling and Simulation of Povetacicept, a Potent Dual BAFF/APRIL Antagonist, to Support Clinical Trials in Multiple Autoantibody- and/or B Cell-Related Diseases

Rupert H. Davies[1], Stacey R. Dillon[1], Russell J. Sanderson[1], Allison Chunyk[1], Mark T. Marino[2], Jeffrey R. Proctor[3], and Stanford L. Peng[1]

[1]Alpine Immune Sciences Inc., Seattle, United States of America; [2]Mark T. Marino Consulting LLC, San Diego, USA and [3]Prosiga Consulting, Vancouver, Canada

Introduction

B cell activating factor (BAFF) and a proliferation-inducing ligand (APRIL) are tumor necrosis factor superfamily (TNFSF) members that bind transmembrane activator and calcium-modulating cyclophilin ligand interactor (TACI), B cell maturation antigen (BCMA), and/or BAFF receptor (BAFF-R) on B cells and together support B-cell development, survival, and differentiation into antibody-secreting cells (ASC).

Therapeutic agents targeting BAFF and/or APRIL have demonstrated promising clinical efficacy and proof-of-concept in systemic lupus erythematosus, lupus nephritis, IgA nephropathy, Sjogren's syndrome, and myasthenia gravis; however, there is still need for more safe and efficacious therapies. Povetacicept (ALPN-303) is an Fc fusion protein of an engineered TACI variant TNFRSF domain (vTD) with enhanced affinity for APRIL and BAFF, which mediates potent inhibitory activity. Povetacicept may therefore significantly improve clinical outcomes in autoantibody- and/or B-cell-related diseases.

In this first-in-human study (NCT05034484), adult healthy volunteers (HV) were randomized into single-ascending-dose cohorts of intravenous (IV) or subcutaneous (SC) povetacicept or placebo. This study provided the PK/PD data for model fitting, and simulations of these models were used to predict repeat dosing of povetacicept and to assess multiple dose regimens.

Figure 1: APRIL and BAFF are Critical Survival and Differentiation Factors Throughout B Cell Development

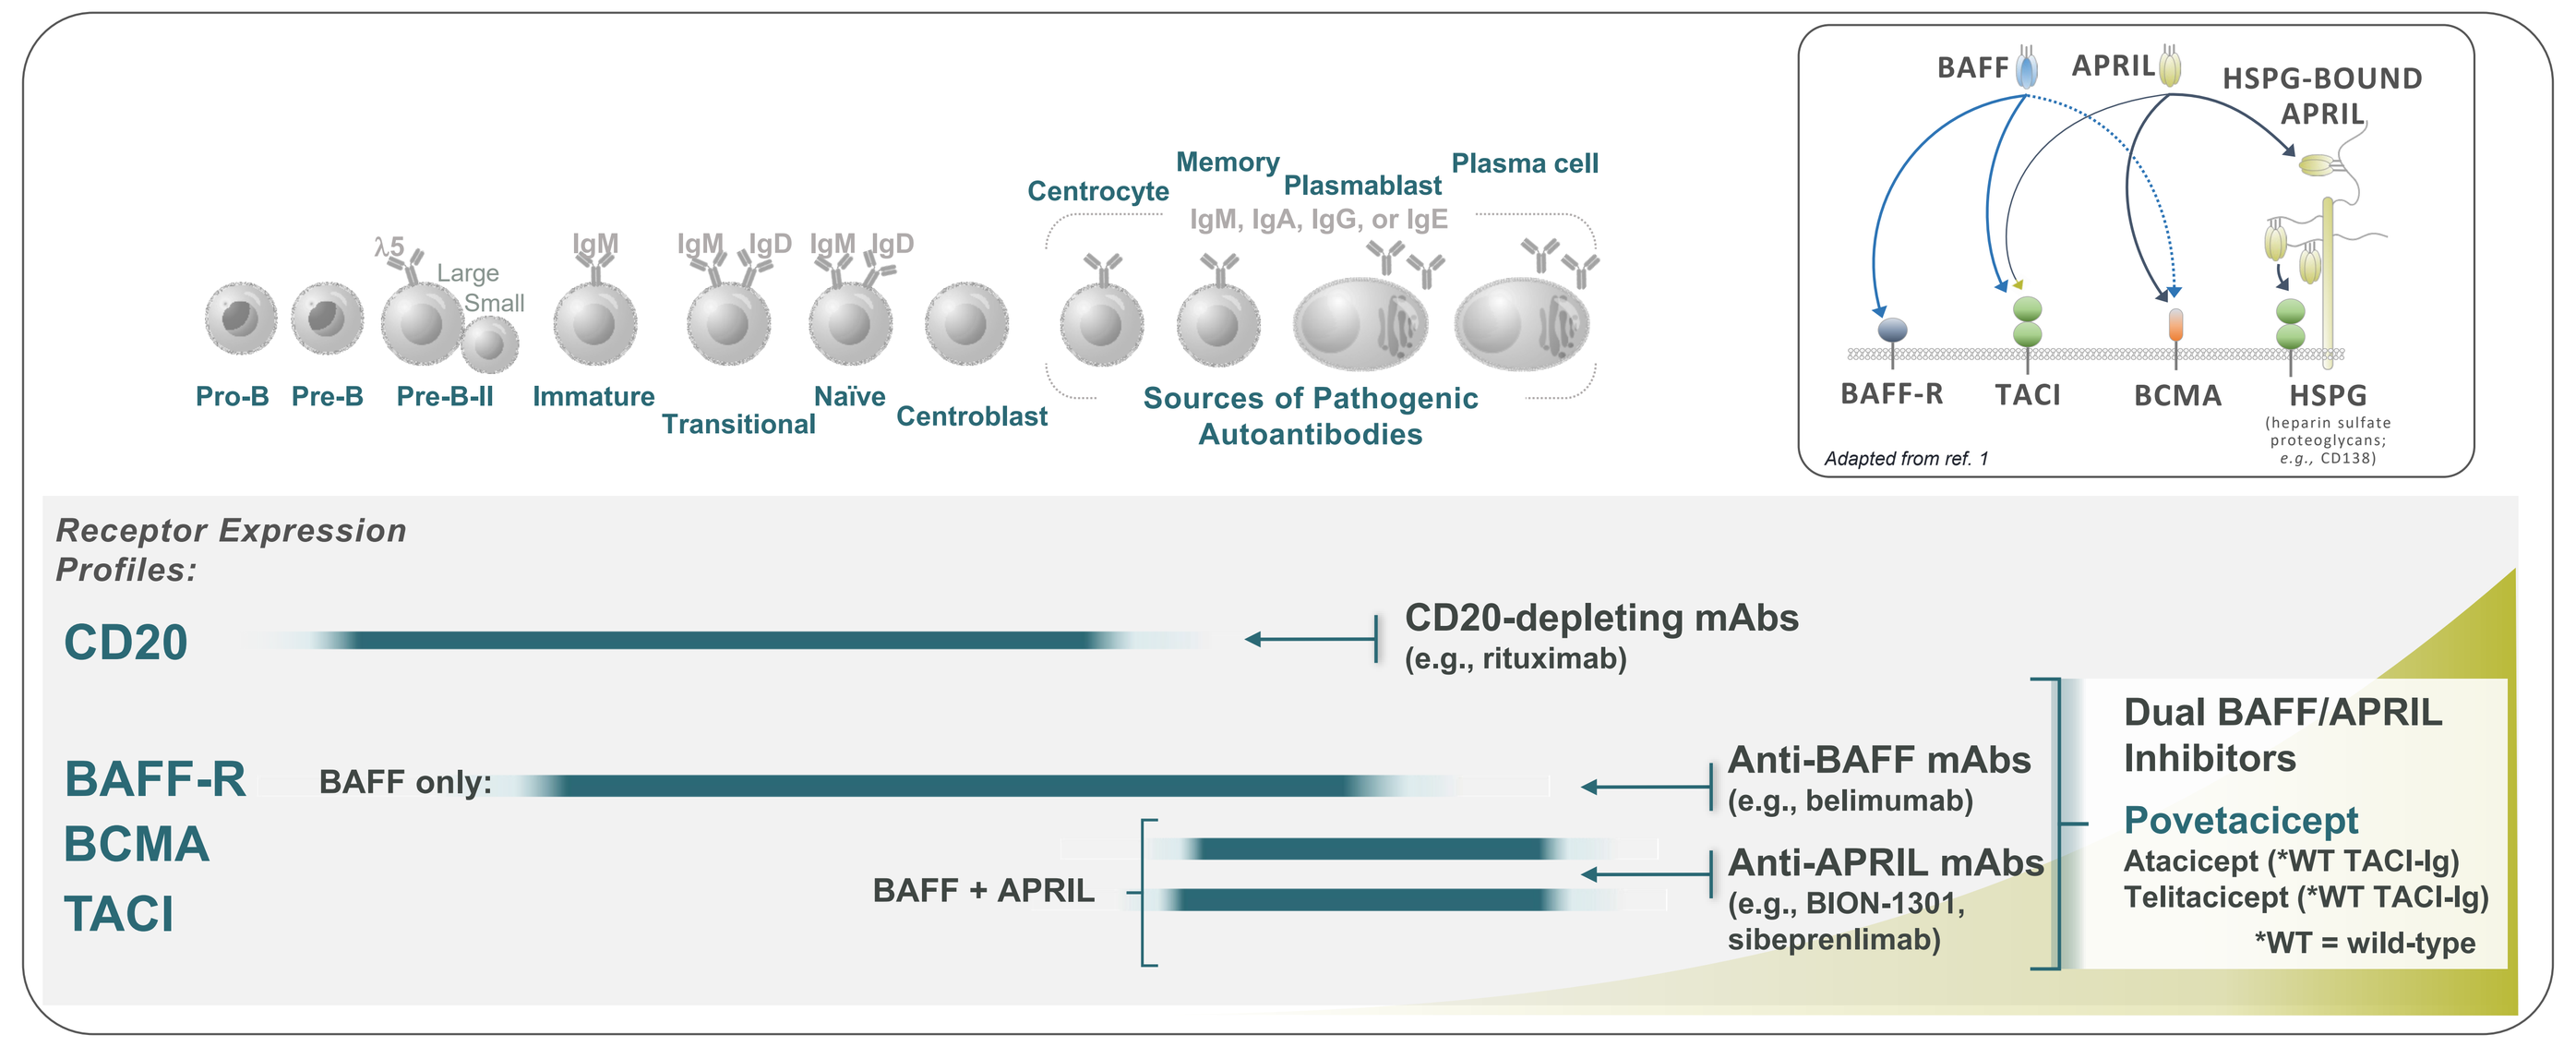
Pro-B
Pre-B
Pre-B-II
Immature
Transitional
Naïve
Centroblast
Centrocyte
Memory
Plasmablast
Plasma cell
IgM, IgA, IgG, or IgE
Sources of Pathogenic Autoantibodies
BAFF
APRIL
HSPG-BOUND APRIL
BAFF-R
TACI
BCMA
HSPG
Adapted from ref. 1
Receptor Expression Profiles:
CD20
BAFF-R
BCMA
TACI
BAFF only:
BAFF + APRIL
CD20-depleting mAbs (e.g., rituximab)
Anti-BAFF mAbs (e.g., belimumab)
Anti-APRIL mAbs (e.g., BION-1301, sibeprenlimab)
Dual BAFF/APRIL Inhibitors
Povetacicept
Atacicept (*WT TACI-Ig)
Telitacicept (*WT TACI-Ig)
*WT = wild-type

Figure 2: Povetacicept is an Enhanced APRIL/BAFF Antagonist that Potently Modulates B Cells and Pathogenic Autoantibodies

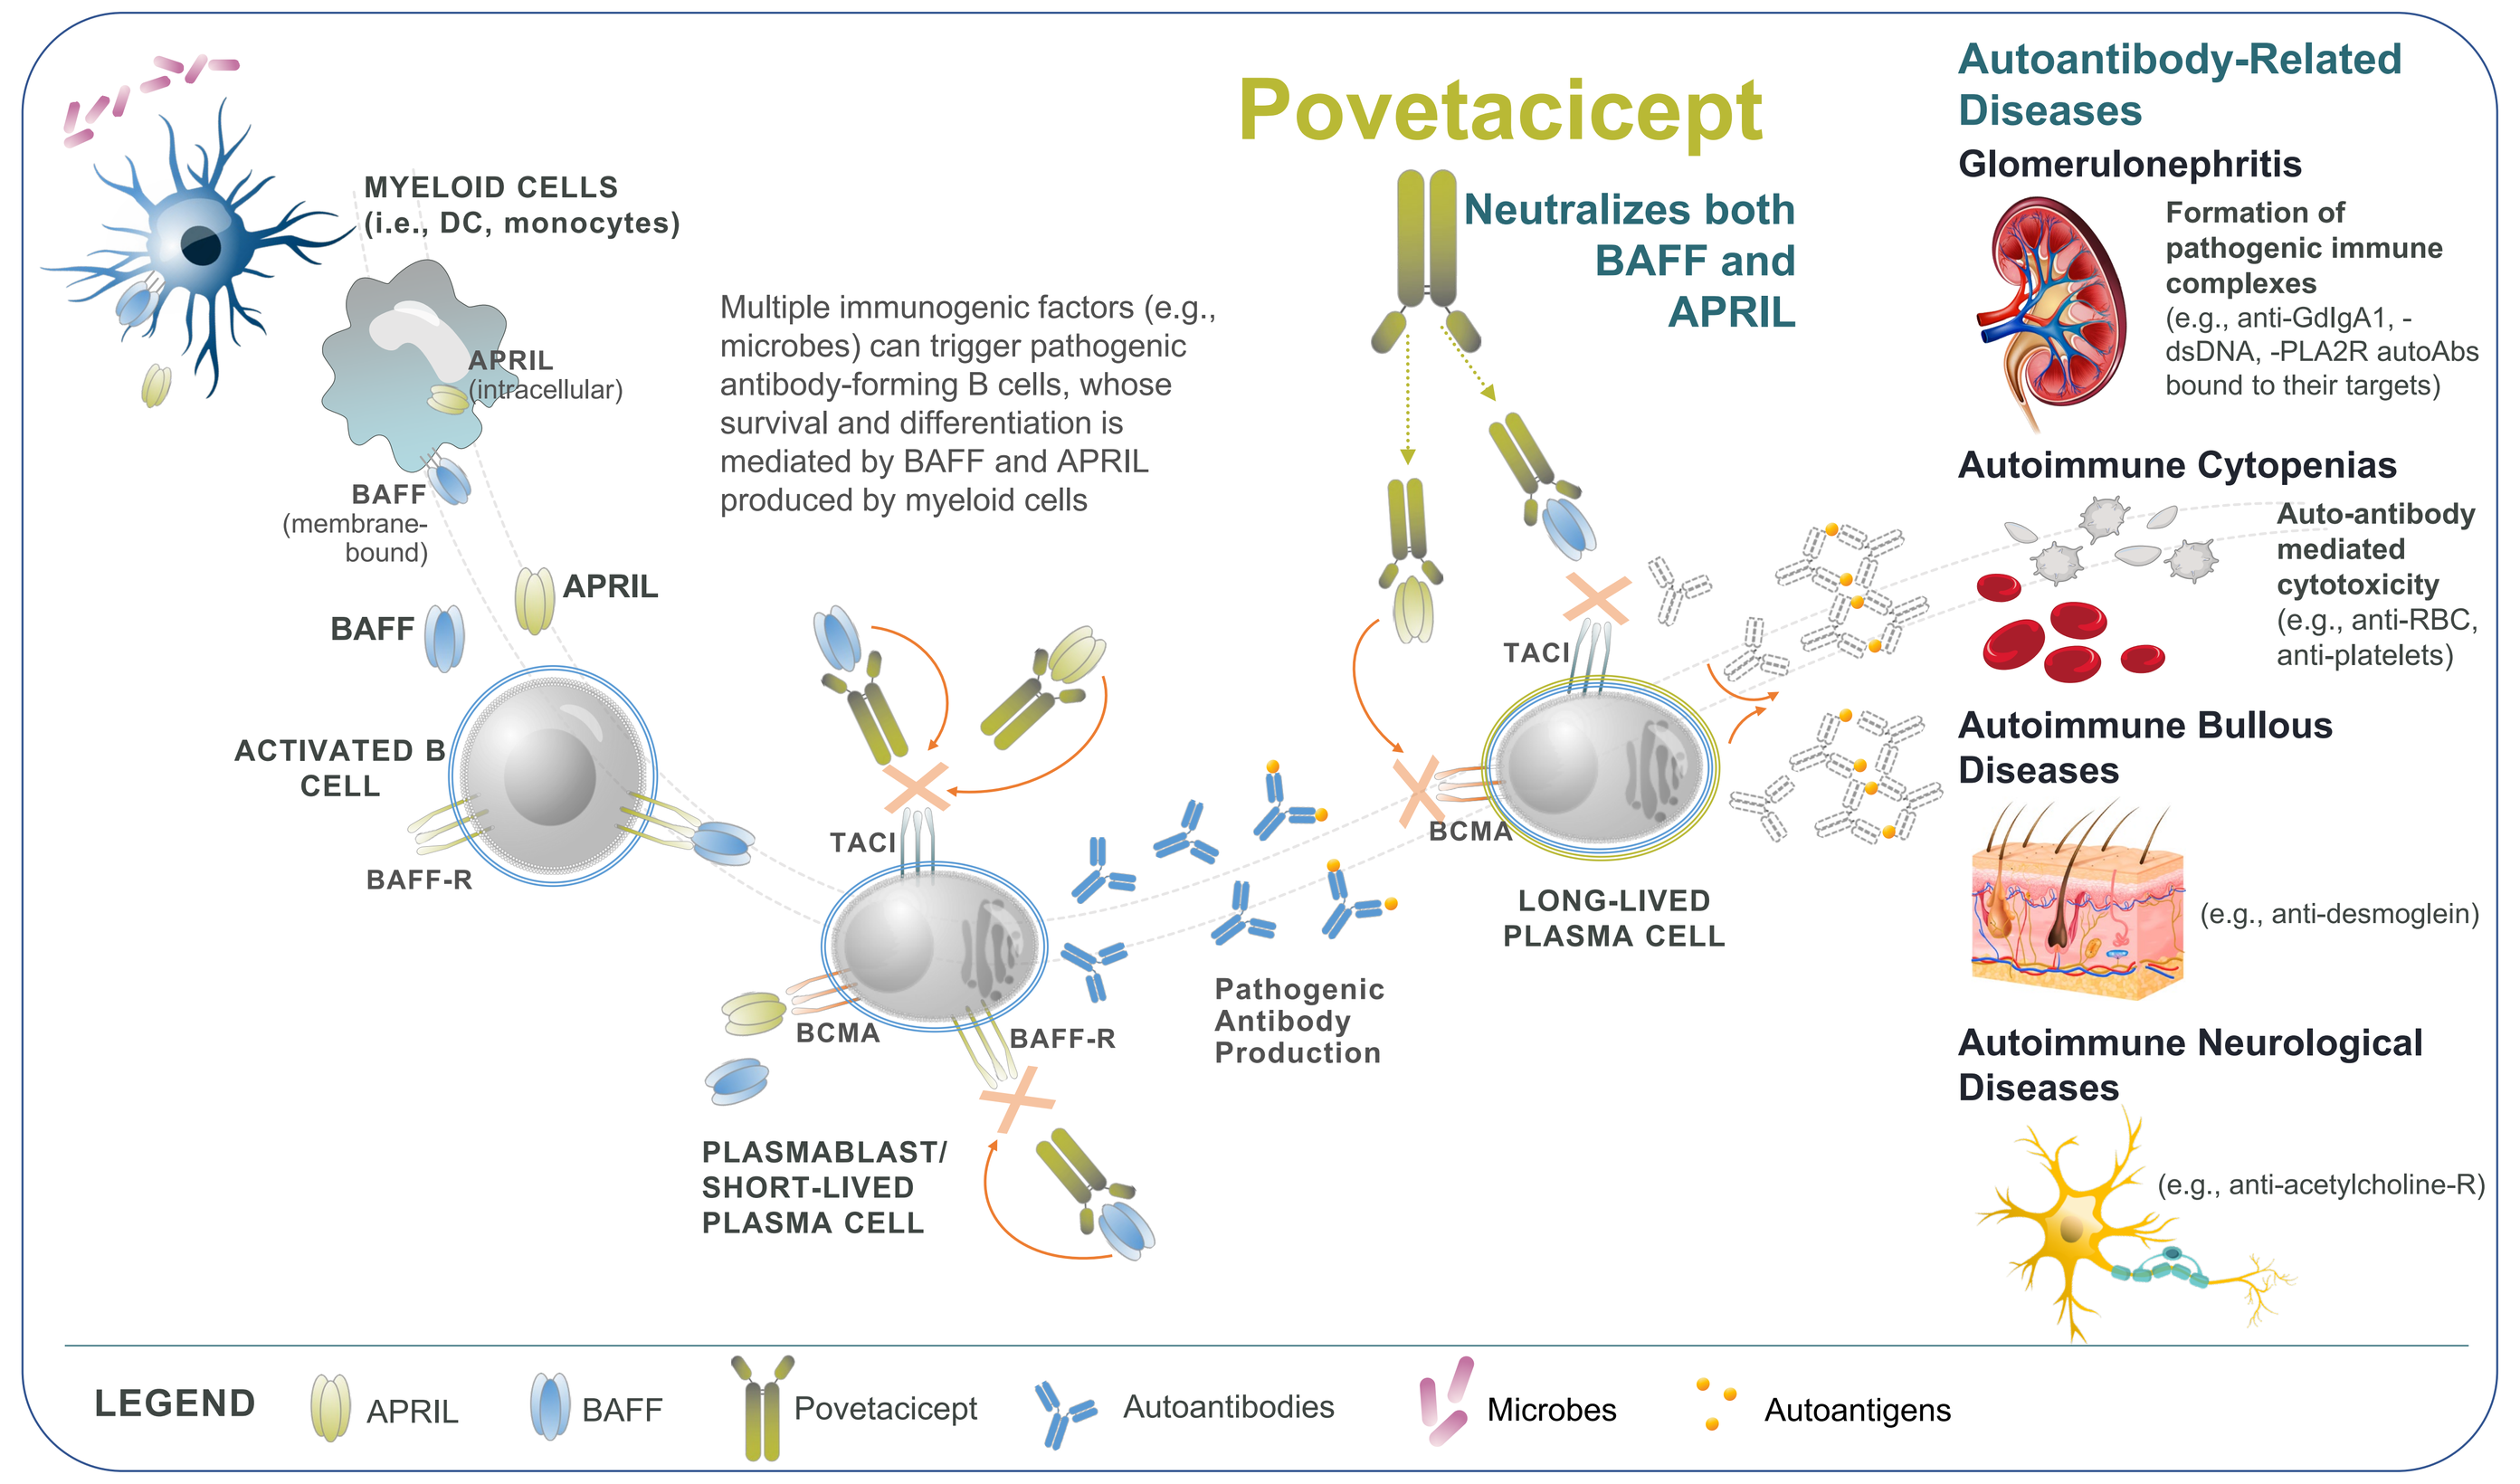
MYELOID CELLS (i.e., DC, monocytes)
APRIL (intracellular)
BAFF (membrane-bound)
APRIL
BAFF
ACTIVATED B CELL
BAFF-R
Multiple immunogenic factors (e.g., microbes) can trigger pathogenic antibody-forming B cells, whose survival and differentiation is mediated by BAFF and APRIL produced by myeloid cells
Povetacicept
Neutralizes both BAFF and APRIL
TACI
BCMA
BAFF-R
PLASMABLAST/ SHORT-LIVED PLASMA CELL
Pathogenic Antibody Production
LONG-LIVED PLASMA CELL
Autoantibody-Related Diseases
Glomerulonephritis
Formation of pathogenic immune complexes (e.g., anti-GdIgA1, -dsDNA, -PLA2R autoAbs bound to their targets)
Autoimmune Cytopenias
Auto-antibody mediated cytotoxicity (e.g., anti-RBC, anti-platelets)
Autoimmune Bullous Diseases
(e.g., anti-desmoglein)
Autoimmune Neurological Diseases
(e.g., anti-acetylcholine-R)
LEGEND
APRIL
BAFF
Povetacicept
Autoantibodies
Microbes
Autoantigens

Figure 3: RUBY-1 Study Design

Study Design

- RUBY-1 (NCT05054484) is a Phase 1 trial evaluating single ascending doses of 2.4 to 960 mg povetacicept administered IV or SC to healthy volunteers
- PK/PD models (direct and indirect) were fit to exposure and PD data.
- Monte Carlo simulations of these models were used to predict the APRIL target coverage and decreases in IgA, IgG, and IgM to estimate the impact of povetacicept in repeat-dose studies.
- Repeat-dose stimulations were performed out to 24 weeks with 16 weeks of recovery

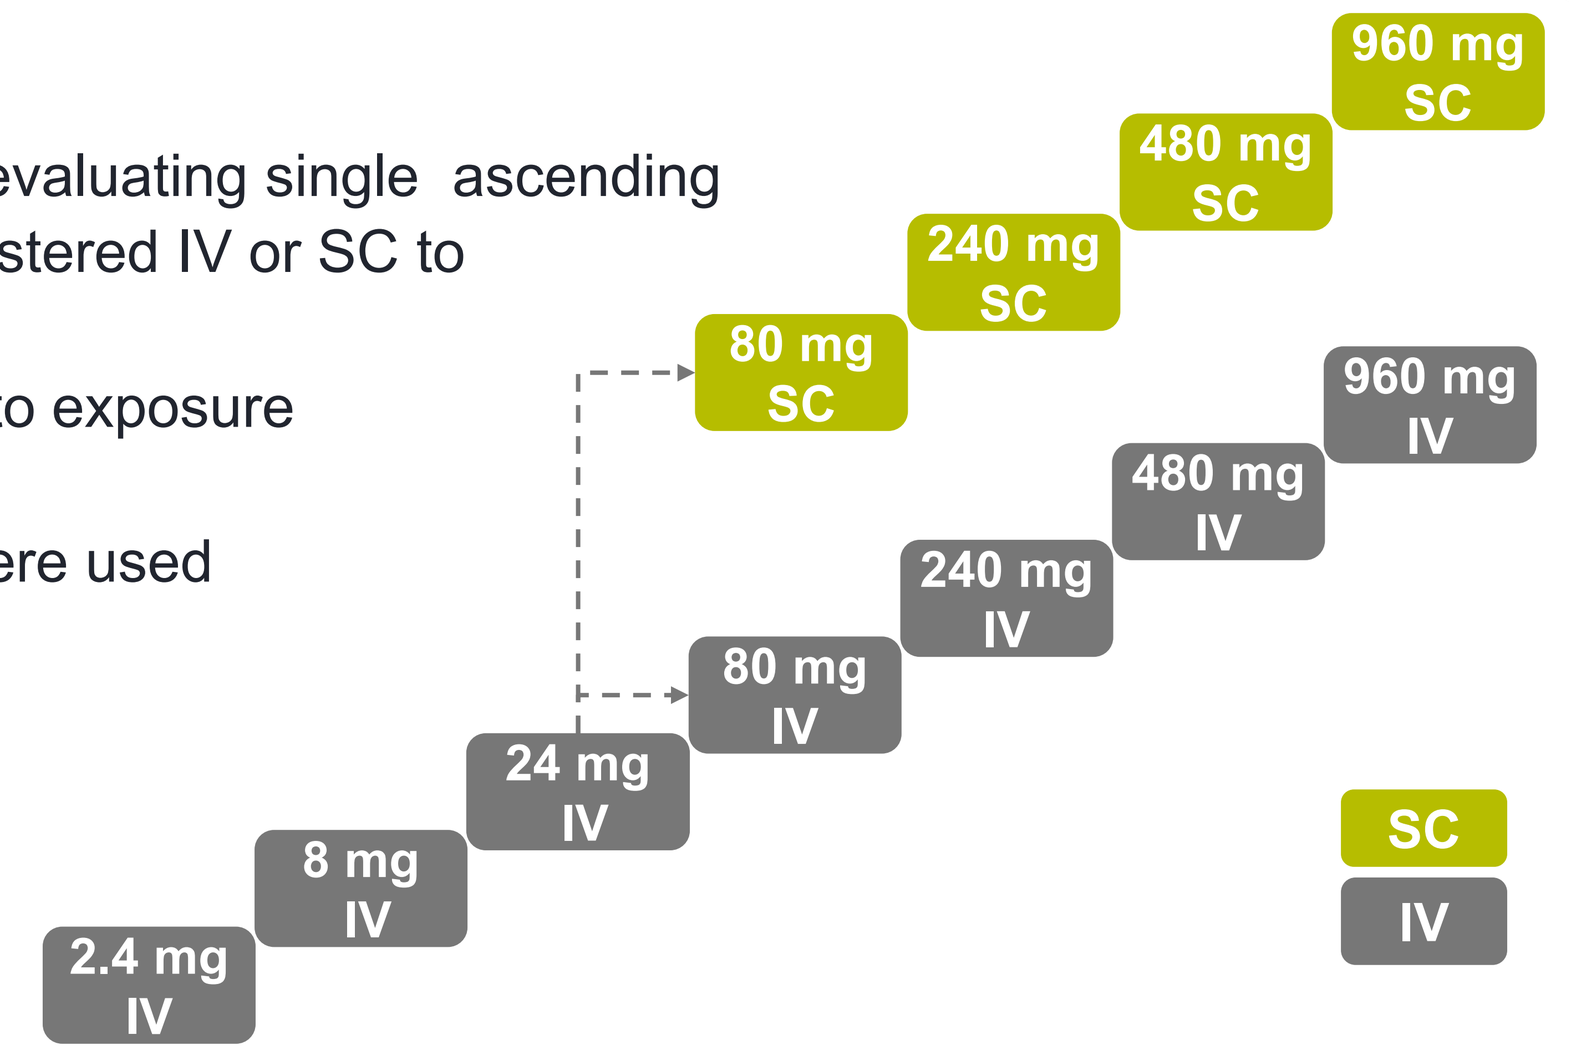
2.4 mg IV
8 mg IV
24 mg IV
80 mg IV
240 mg IV
480 mg IV
960 mg IV
80 mg SC
240 mg SC
480 mg SC
960 mg SC
SC
IV

Figure 4: Povetacicept PK Model Structure, Model Diagnostic Plots and Visual Predictive Assessment

PK Model Structure

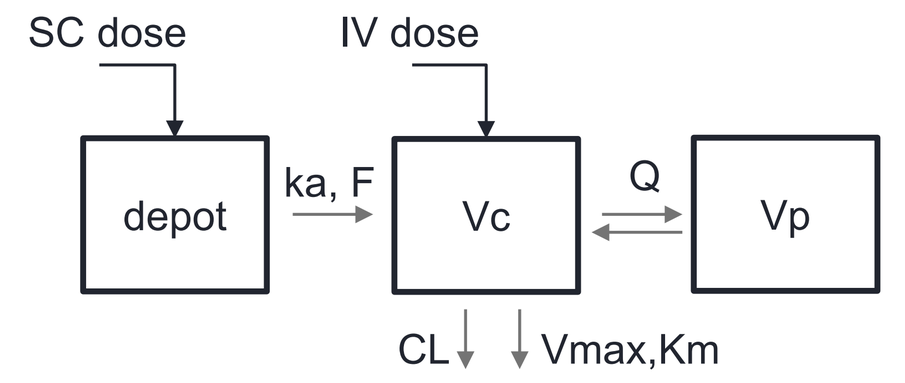
SC dose
IV dose
depot
ka, F
Vc
Q
Vp
CL
Vmax,Km

- Two compartment model with first order absorption
- Parallel linear and non-linear (Michaelis- Menten) clearance
- Inter-individual variability estimated on CL, Vc
- All parameters estimated with RSE < 40%
- < 20% shrinkage in PK model

PK Model Diagnostic Plots

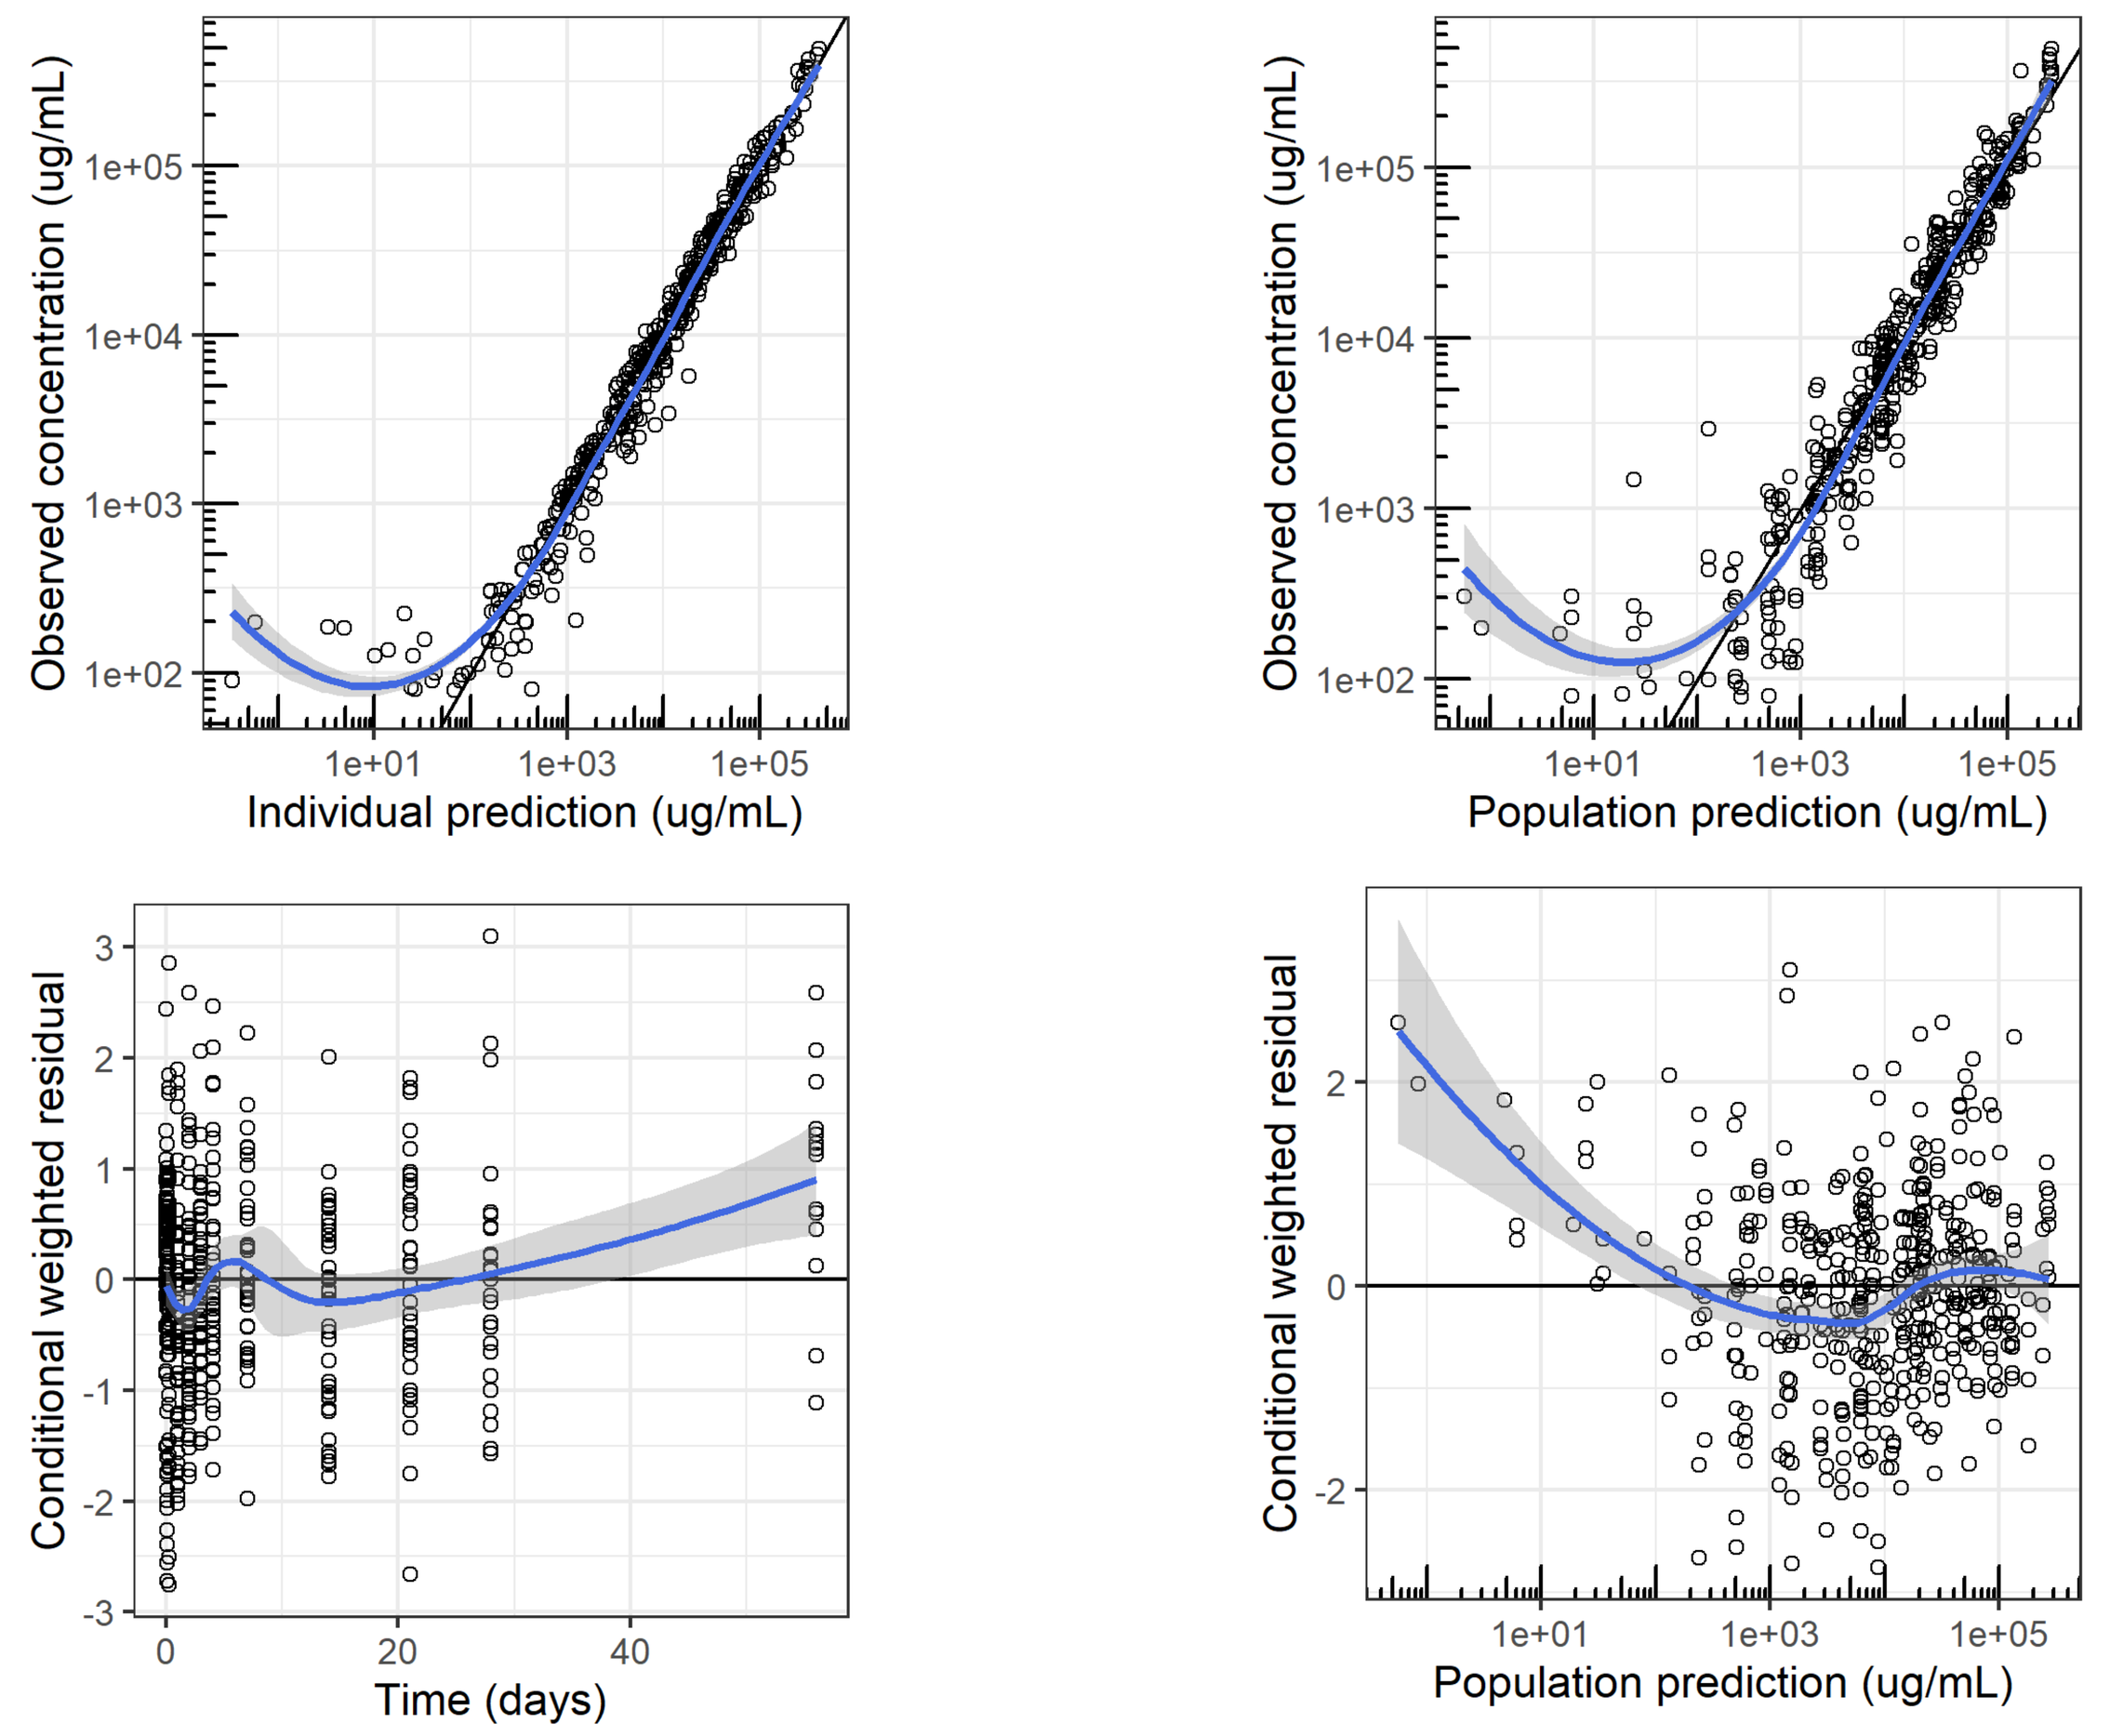
Observed concentration (ug/mL)
Individual prediction (ug/mL)
Population prediction (ug/mL)
Conditional weighted residual
Time (days)

PK Visual Predictive Assessment

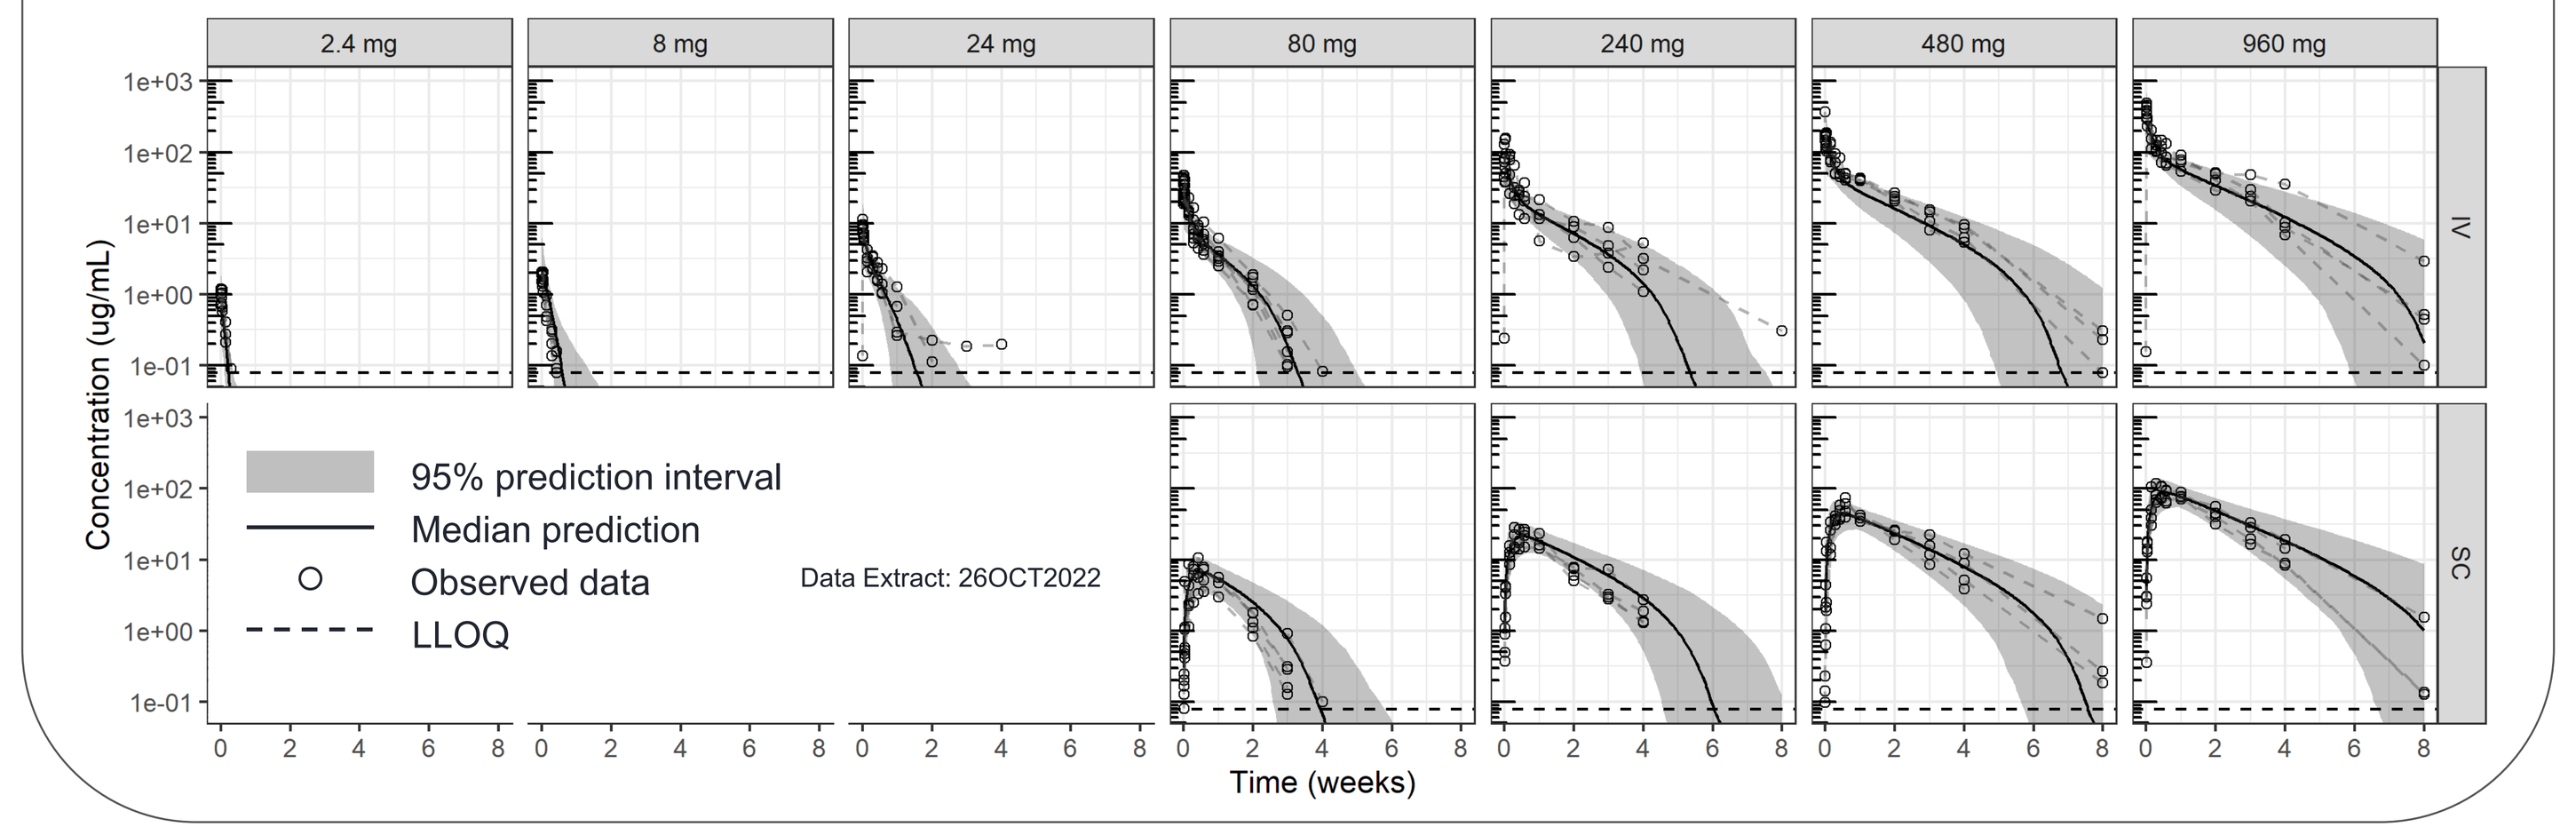
2.4 mg
8 mg
24 mg
80 mg
240 mg
480 mg
960 mg
IV
SC
Concentration (ug/mL)
Time (weeks)
95% prediction interval
Median prediction
Observed data
LLOQ
Data Extract: 26OCT2022

Figure 5: Povetacicept PK Simulation

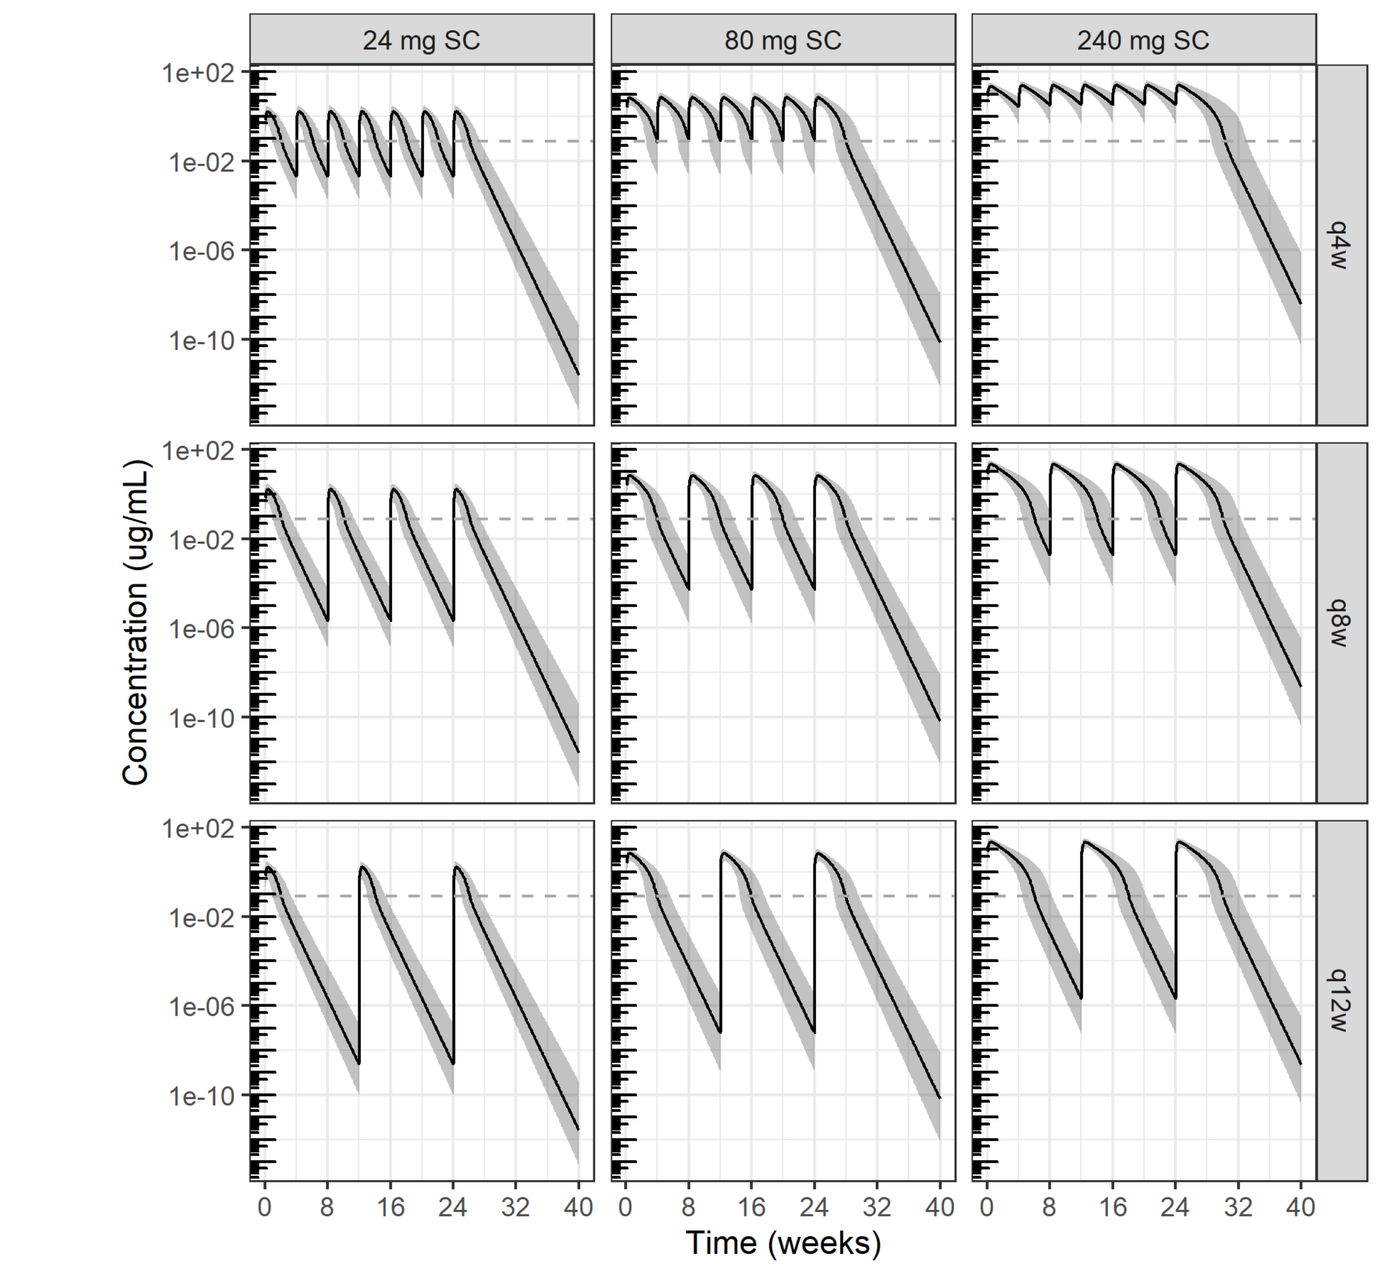
24 mg SC
80 mg SC
240 mg SC
q4w
q8w
q12w
Concentration (ug/mL)
Time (weeks)
95% prediction interval
Median prediction
LLOQ

- Based on NCA, the half-life of povetacicept at 80 and 240 mg SC is 3.7 days and 7.4 days, respectively.
- Repeat dosing of povetacicept, at dose intervals of 4 weeks or more, is predicted to have little to no accumulation
- With the exception of the 80 and 240 mg Q4W dosing regimens, non-linear clearance dominates all dose regimens simulated
- Steady state is reached by the second dose due to the rapid clearance

Figure 6: Povetacicept PK/PD Model Structures and Visual Predictive Assessments

PK/PD Model Structures

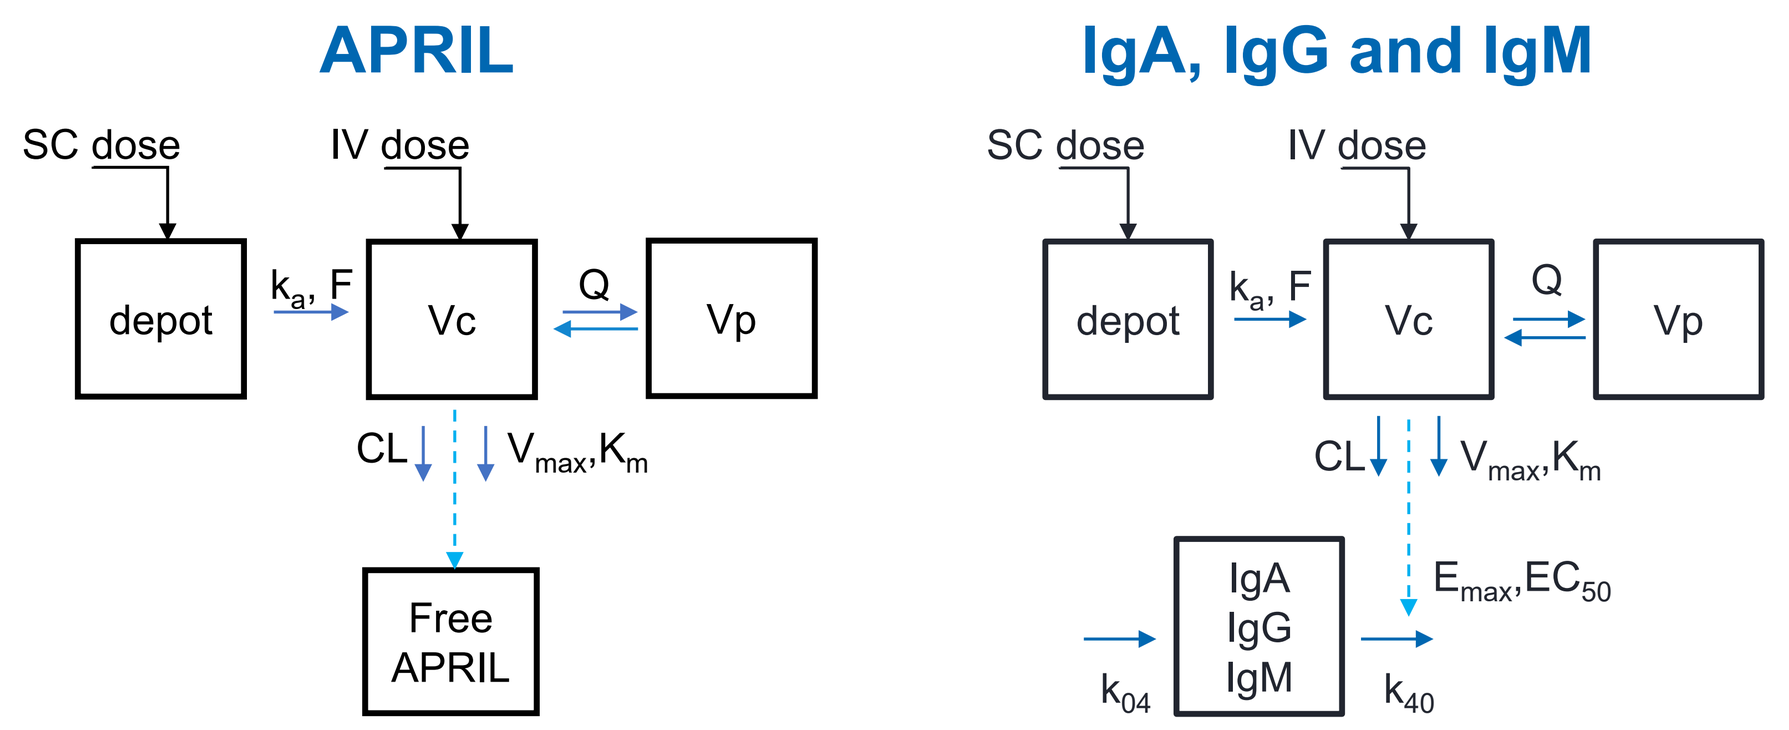
APRIL
IgA, IgG and IgM
SC dose
IV dose
depot
Vc
Vp
Q
CL
Free APRIL
IgA IgG IgM

- APRIL, IgA, IgG and IgM PK/PD models were fitted individually and had specific parameters
- Free APRIL concentrations are best described with a direct model
- IgA, IgG and IgM concentrations are best described by an indirect model with a saturable effect

PK/PD Visual Predictive Assessments

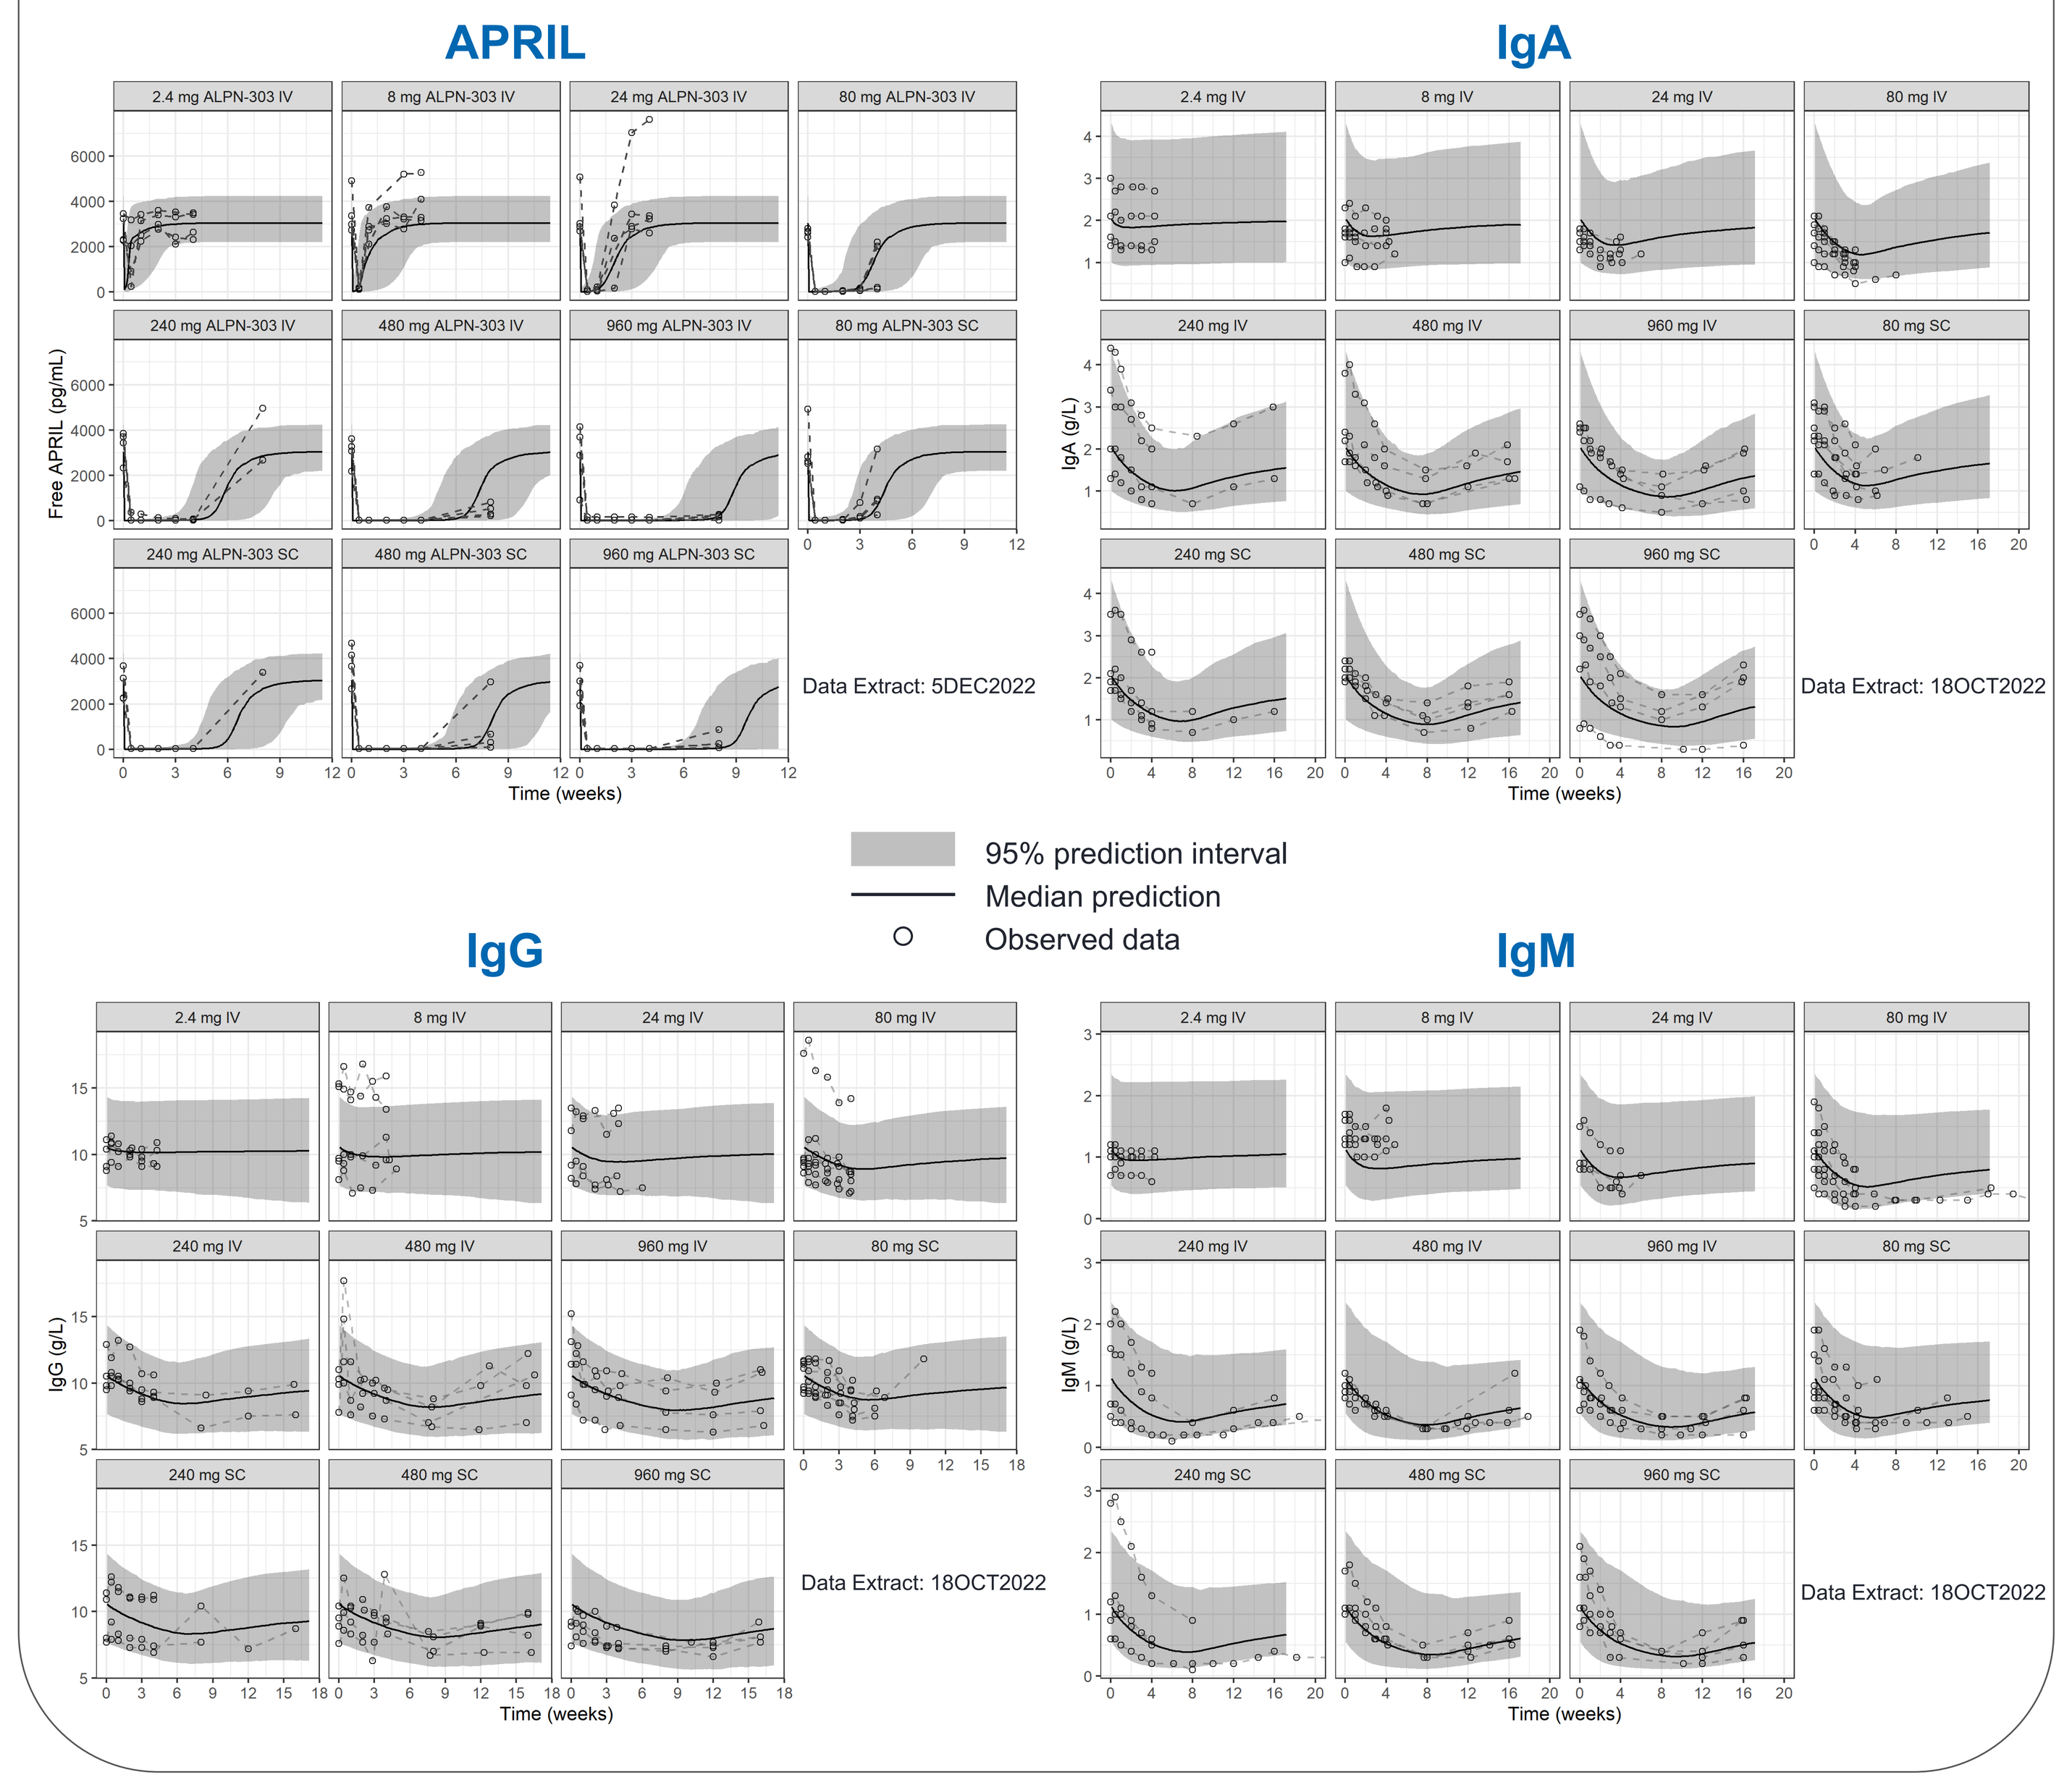
APRIL
IgA
IgG
IgM
Free APRIL (pg/mL)
Time (weeks)
Data Extract: 5DEC2022
Data Extract: 18OCT2022
95% prediction interval
Median prediction
Observed data

Figure 7: Povetacicept PK/PD Simulations

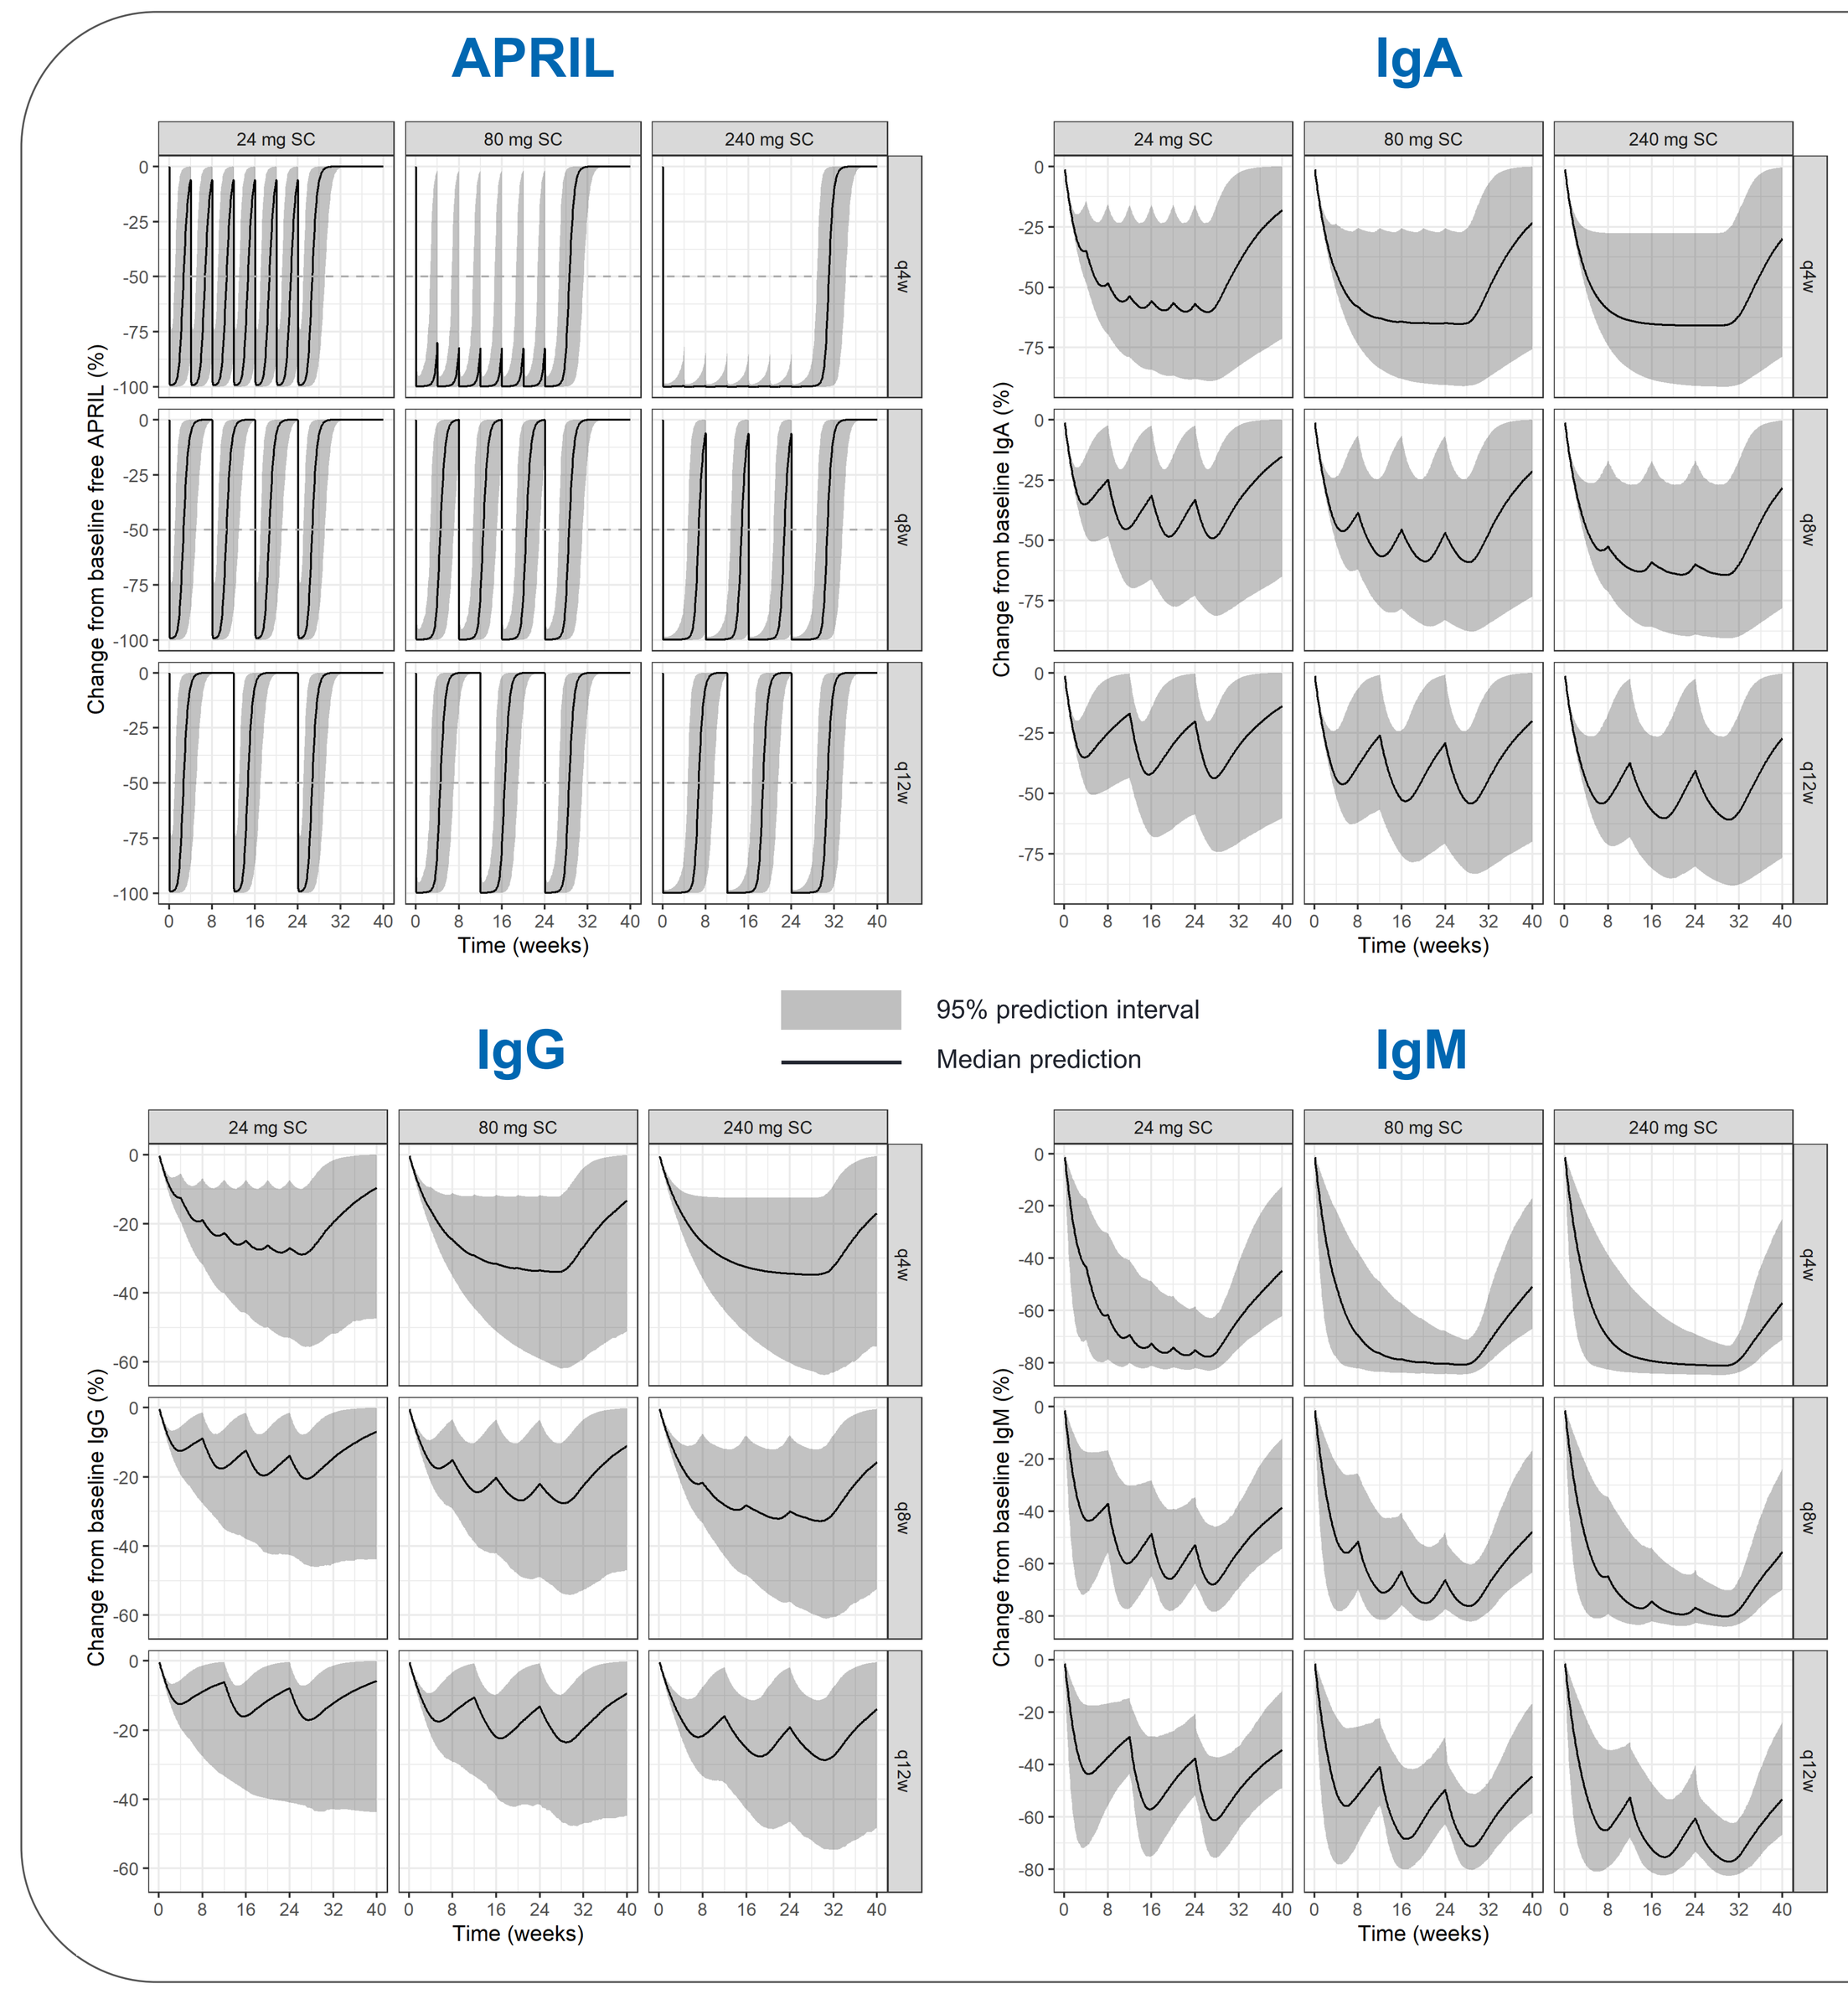
APRIL
IgA
IgG
IgM
24 mg SC
80 mg SC
240 mg SC
q4w
q8w
q12w
Time (weeks)
95% prediction interval
Median prediction

- Simulations show that APRIL is quickly bound by all dose regimens within <1 day and for dose regimens of 80 and 240 mg Q4W, median APRIL recovers to baseline levels within 8 to 10 weeks, respectively.
- APRIL simulations predict that median patient dosed 80 and 240 mg SC Q4W will have 75% and >100% reductions in APRIL for entire dosing period.
- IgA, IgG and IgM simulations show the following dose regimens have saturated reductions of IgA, IgG, and IgM:
 - 80 mg SC Q4W
 - 240 mg SC Q4W and Q8W
- Median IgA, IgG and IgM reach nadir by 8-12 weeks after repeat dosing of povetacicept.

Figure 8: Summary of Target Coverage in Simulations

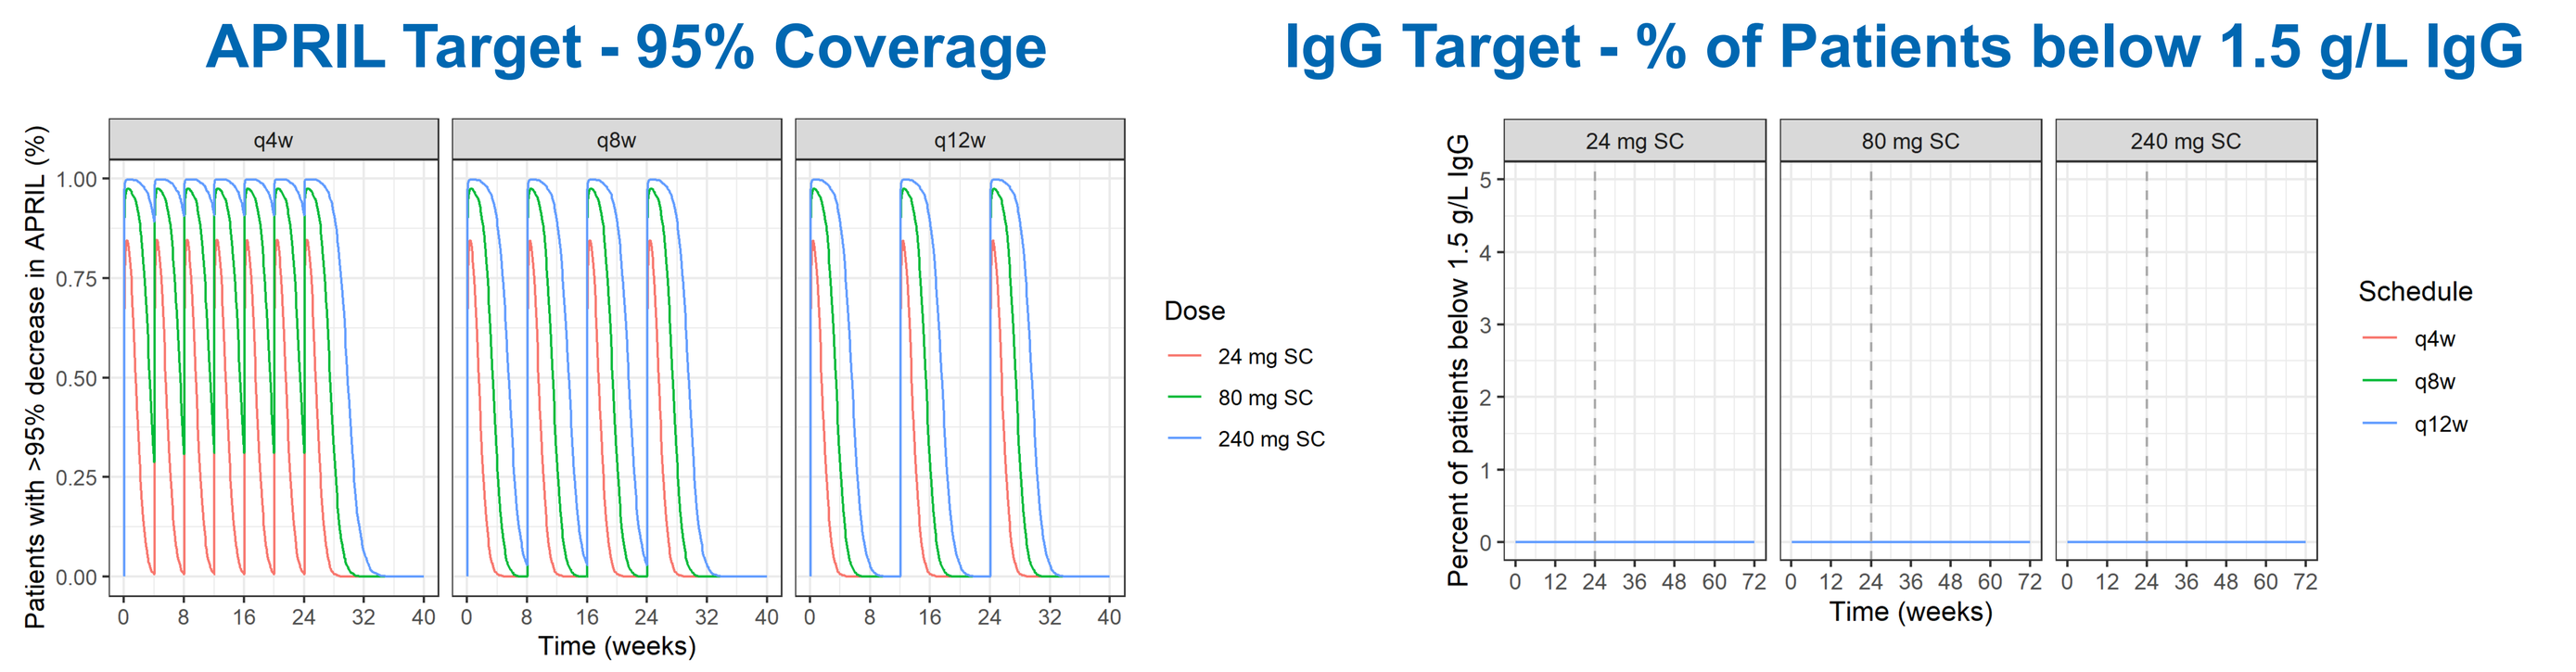
APRIL Target - 95% Coverage
IgG Target - % of Patients below 1.5 g/L IgG
q4w
q8w
q12w
Dose
24 mg SC
80 mg SC
240 mg SC
Schedule
Time (weeks)

- APRIL simulations show that dose regimens of 80 and 240 mg SC Q4W have greater than 75% and 95% target coverage for 3 and 4 weeks at steady state, respectively.
- IgG simulations predict that dose regimens of 80 and 240 mg SC Q4W have less than 1% of patients will have reductions in IgG that would result in hypogammaglobulinemia (<1.5 g/L IgG).
- These dose regimens of 80 and 240 mg SC Q4W are anticipated to provide adequate APRIL target coverage while minimizing risk of hypogammaglobulinemia.

Summary and Conclusions

- Povetacicept exposure is best described by a PK model with parallel linear and nonlinear clearance.
- APRIL and immunoglobulin concentrations are best described by direct and indirect models with a saturable effect, respectively.
- Dose regimens of 80 and 240 mg SC Q4W are anticipated to provide adequate APRIL target coverage while minimizing risk of hypogammaglobulinemia.
- Simulations of these PK/PD models support investigating the dose regimens of 80-240 mg SC every 4 weeks in future studies.

References

1. Sakai *et al.* Clin Microbiol Rev. 2017. Oct; 30(4): 991-1014.
2. Hébert and Joly. Immunotherapy. 2018 Jan; 10(1): 27-37.

Acknowledgements

We thank Jeff Proctor (Prosiga Consulting) and Jennifer Austin (Biocraft Studio).

AlpineImmuneSciences.com
@AlpineImmuneSci

© 2023 Alpine Immune Sciences, Inc. All rights reserved.